287

ONE THIRD OF THE PORTUGUESE POPULATION (20-79 YEARS) HAS DIABETES OR "PRE-DIABETES" (PREVADIAB-Study)

Diabetes Prevalence Study in Portugal

Authors: L. Gardete-Correia[1], J. M. Boavida[3,4], J. F. Raposo[1], A. C. Mesquita[1], C. Fona[1], R. Carvalho[1] and S. Massano-Cardoso[2],

[1]Portuguese Diabetes Association (APDP), Lisbon, Portugal, [2]Hygiene and Social Medicine Institute, Coimbra, Portugal, [3]Portuguese Diabetes Program, Directorate General of Health, Lisbon, Portugal, [4]Portuguese Society of Diabetology, Lisbon, Portugal.

OBJECTIVES

The objectives of this study were to determine, for the Portuguese population aged between 20 and 79 years:

- The national and comparative standardized prevalences of type 2 diabetes and pre-diabetes defined as Impaired Fasting Glucose (IFG) levels and Impaired Glucose Tolerance (IGT).
- The prevalence of cardiovascular risk factors (hypertension, dyslipidaemia).

RESEARCH DESIGN AND METHODS

- Considering the number of inhabitants between 20 and 79 years (7,657,529), statistical units were selected that were regionally and nationally representative.
- The total sample in the study comprised 5167 subjects.
- Analyses were performed on all non-diabetic subjects using WHO referenced diagnostic criteria.
- An OGTT was performed (except on previously known diabetes) using WHO referenced diagnostic criteria.
- Blood pressure measurements were recorded in the sitting position using an automatic oscillometric method.
- LDL cholesterol determinations were performed by Olympus AV 640 autoanalyser.

STATISTICAL TREATMENT

- All the analyses were conducted using JMP 6.0.3. Statistical Discovery from SAS.
- The national data were standardized for the Portuguese population aged 20 to 79 years, using the 2001 Census and the comparative prevalence was calculated using the percentile distribution per 5-year age group of the world population.

RESULTS

- National diabetes prevalence is 11.7%, (95% CI 10.8 - 12.6%) with a significant difference between men: 14.2% (95% CI 12.5 - 15.5%); and women: 9.5% (95% CI 8.5 to 10.6%).
- While 6.6% of the subjects had previously been diagnosed with diabetes, 5.1% were undiagnosed.
- Comparative prevalence found was 9.8% (5.3% previously diagnosed and 4.4% undiagnosed).
- By age groups, the results show that 2.4% of the population aged between 20 and 39 years, 12.6% of people from 40 to 59 and 26.3% of people aged between 60 and 79 have diabetes. 23.2% have IGF and/or IGT.

DIABETES PREVALENCE IN PORTUGAL - 2009

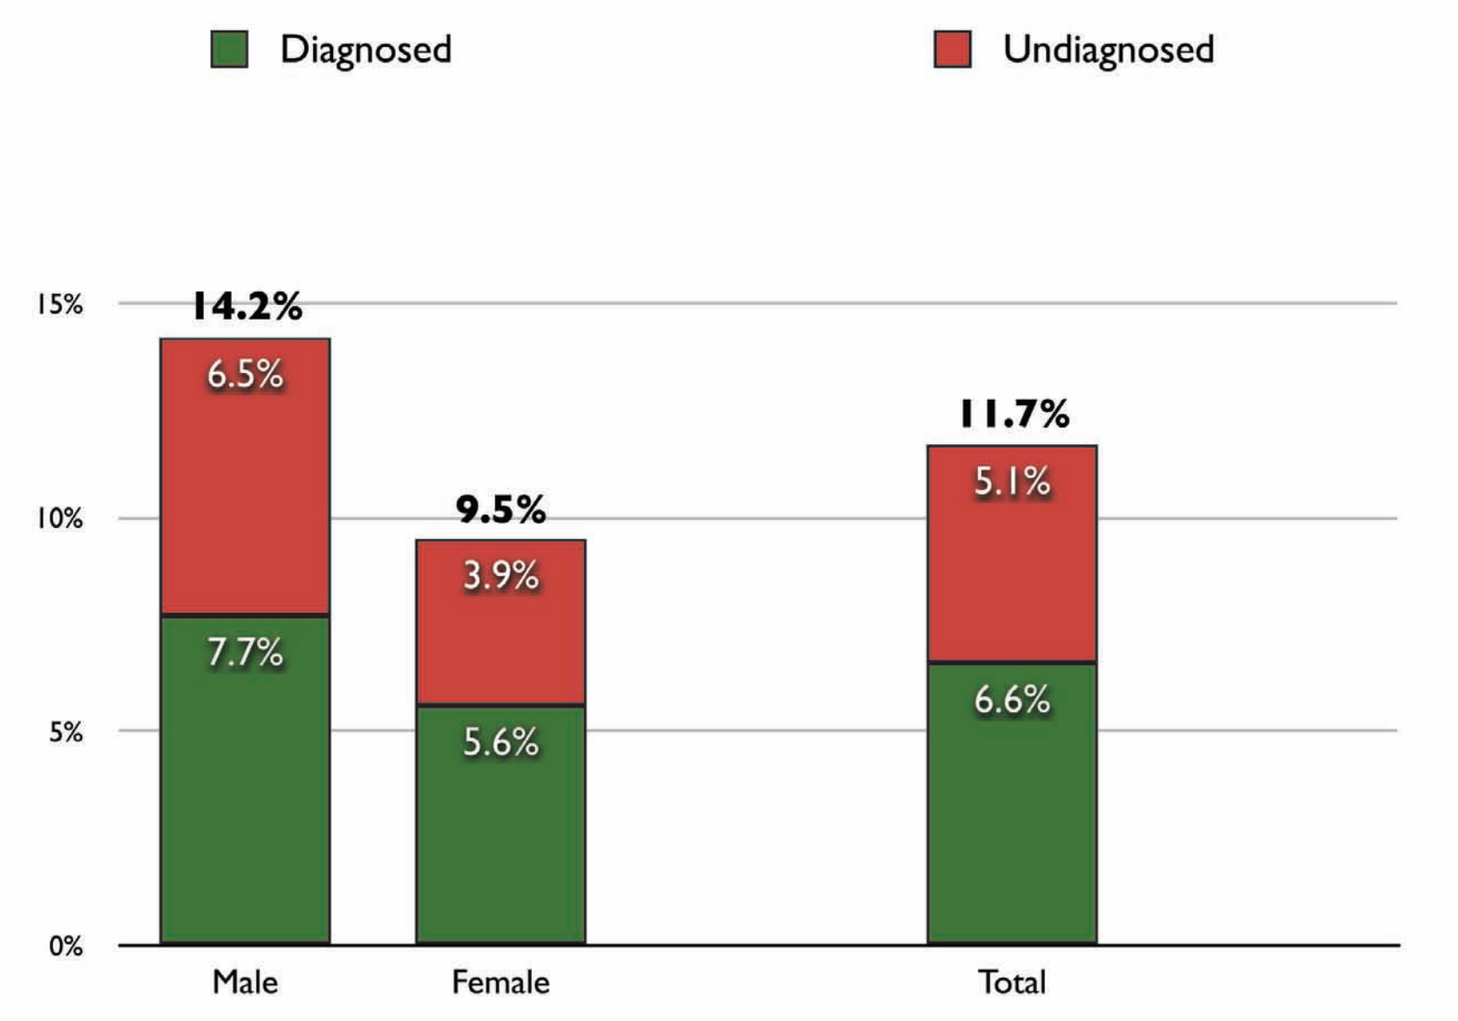
Diagnosed
Undiagnosed
15%
10%
5%
0%
14.2%
6.5%
7.7%
9.5%
3.9%
5.6%
11.7%
5.1%
6.6%
Male
Female
Total

Total prevalence of diabetes (previously diagnosed and undiagnosed) was 11.7% (CI 95% 10.8 to 12.6%) in persons aged 20 to 79 years, with 14.2% in males (CI 95% 12.5 to 15.5%) and 9.5% in females (CI 95% 8.5 to 10.6%)

DIABETES PREVALENCE BY AGE GROUPS Diagnosed/Undiagnosed

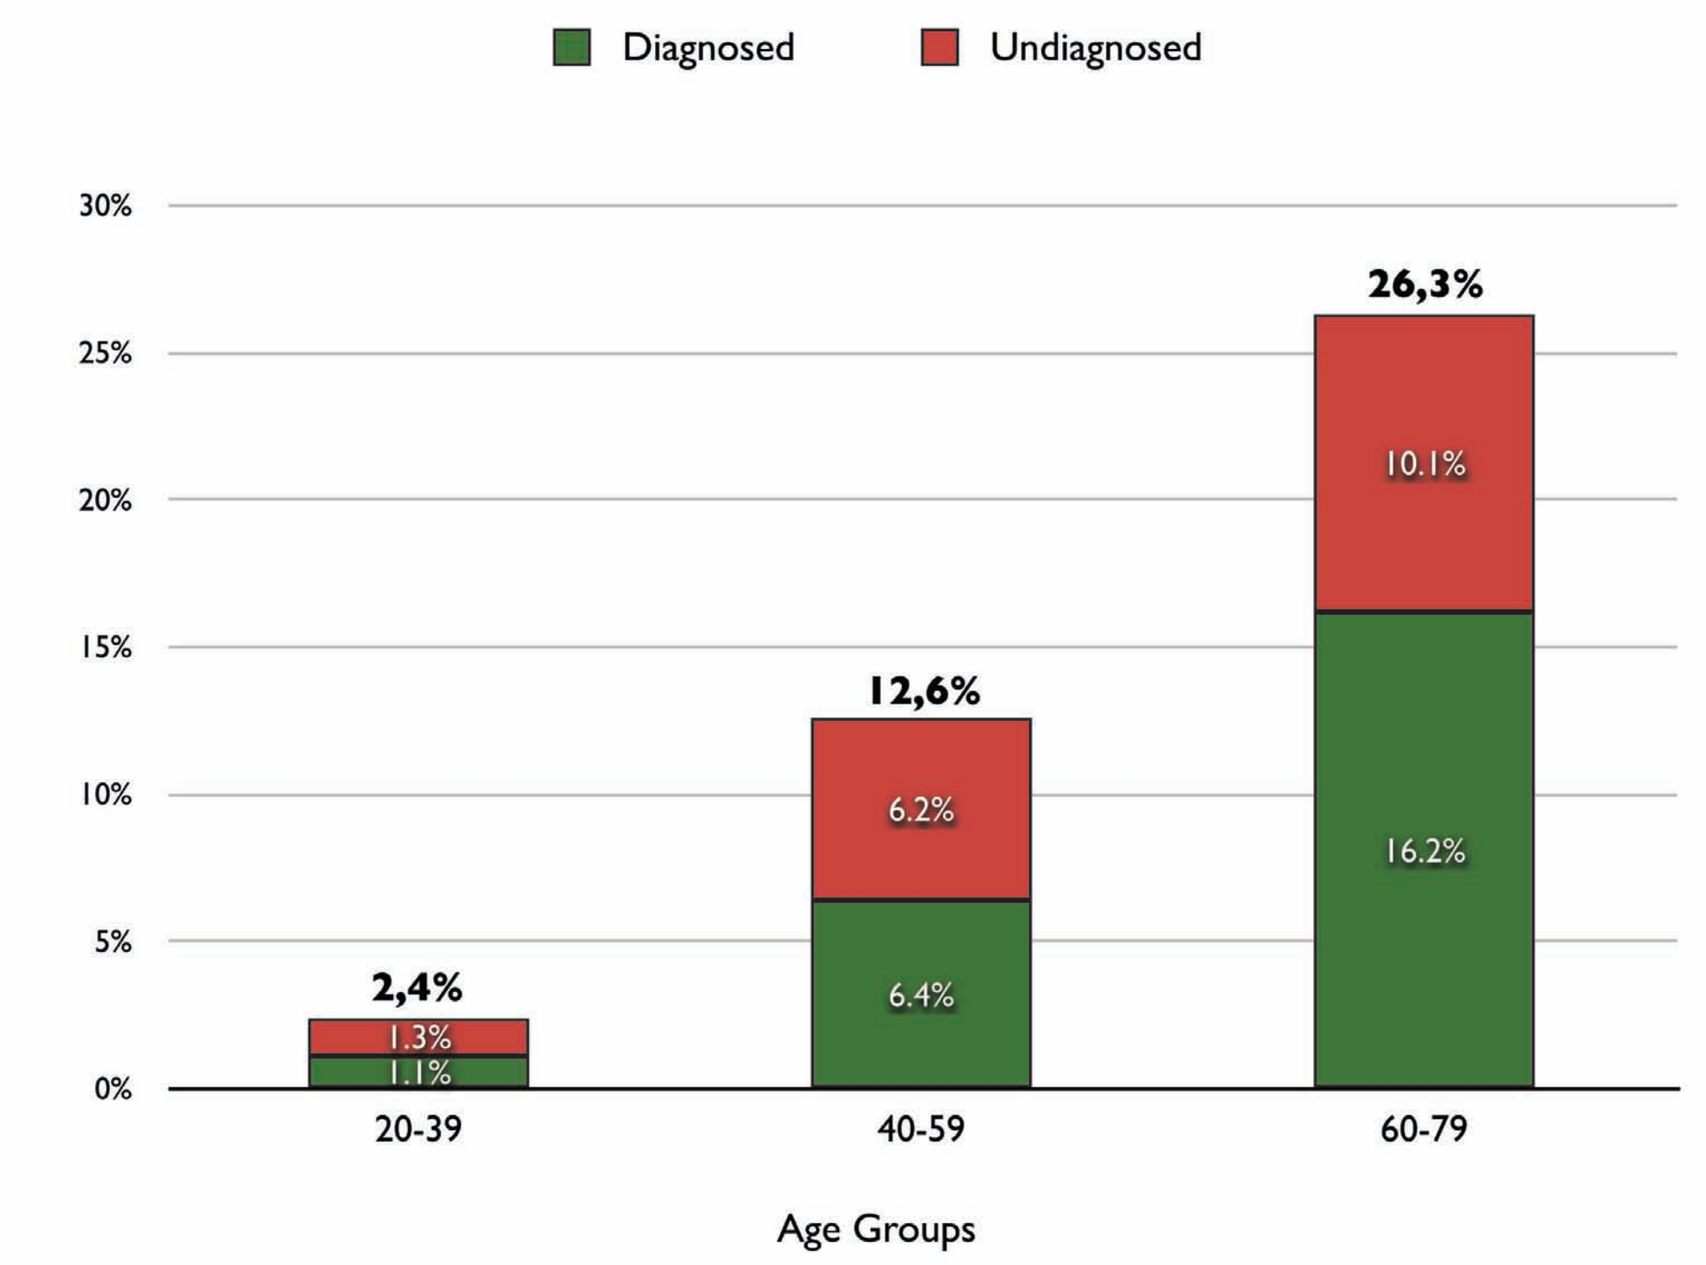
Diagnosed
Undiagnosed
30%
25%
20%
15%
10%
5%
0%
2,4%
1.3%
1.1%
12,6%
6.2%
6.4%
26,3%
10.1%
16.2%
20-39
40-59
60-79
Age Groups

PREVALENCE OF UNDIAGNOSED DIABETES Detected by fasting glycaemia and after a tolerance test (OGTT)

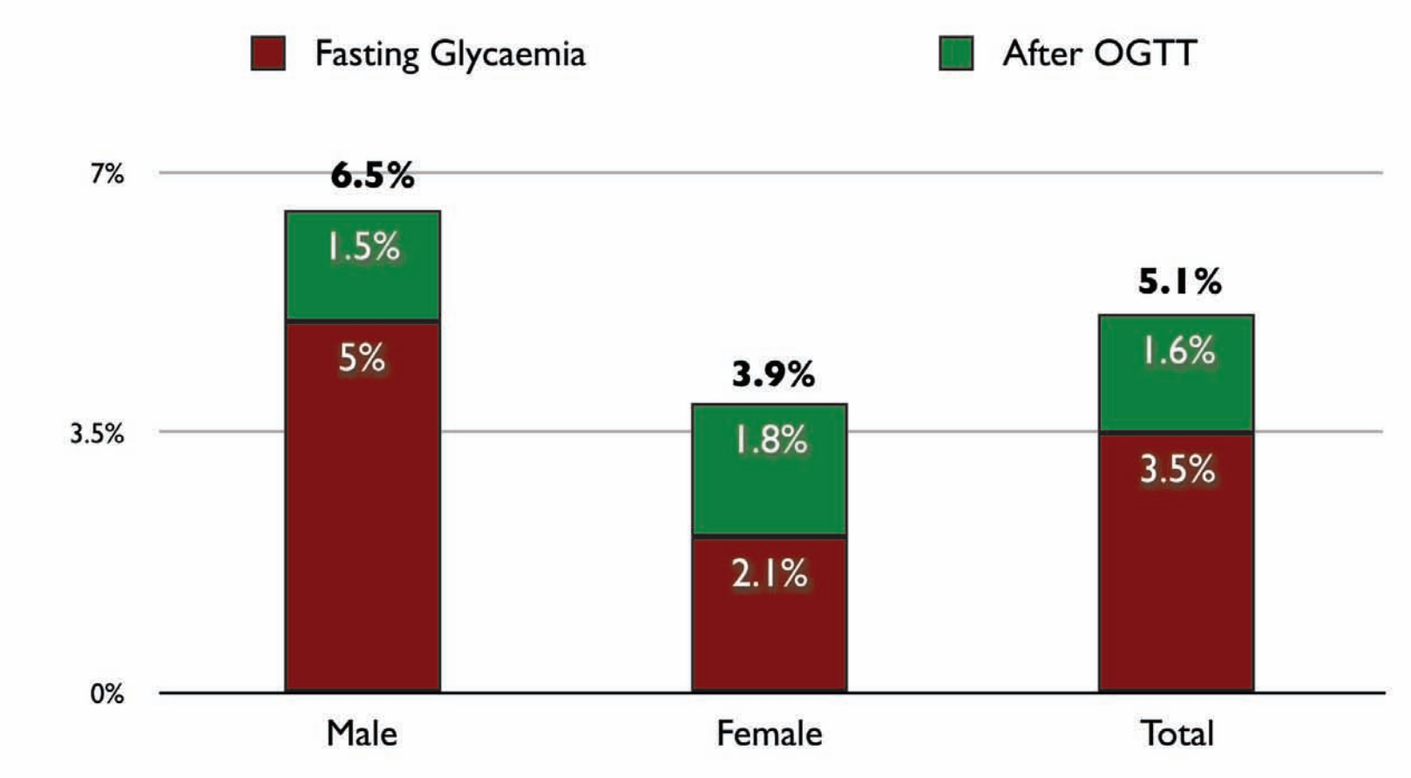
Fasting Glycaemia
After OGTT
7%
3.5%
0%
6.5%
1.5%
5%
3.9%
1.8%
2.1%
5.1%
1.6%
3.5%
Male
Female
Total

Diabetes diagnosed by fasting glycaemia and after a tolerance test (OGTT) – The prevalence of undiagnosed diabetes detected using just fasting glycaemia was 3.5% (CI 95% 2.96 to 4.04%) for both sexes; the figure rose to 5.1% (CI 95% 4.5 to 5.7%) after the tolerance test.

"PRE-DIABETES" BY AGE GROUP

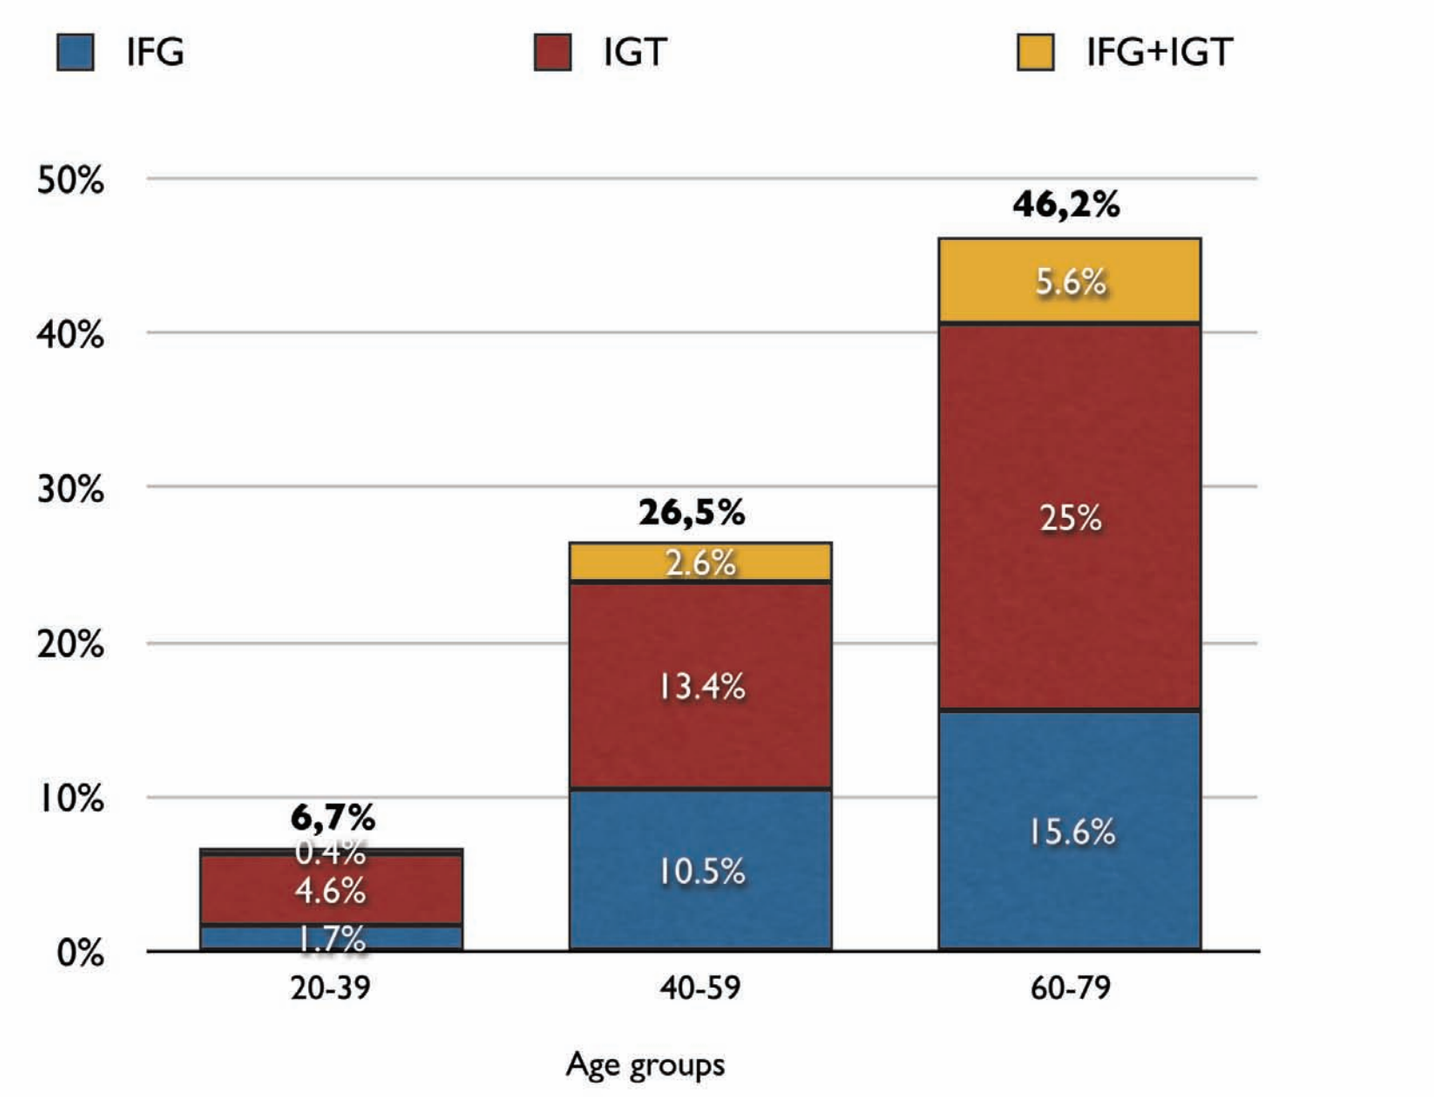
IFG
IGT
IFG+IGT
50%
40%
30%
20%
10%
0%
6,7%
0.4%
4.6%
1.7%
26,5%
2.6%
13.4%
10.5%
46,2%
5.6%
25%
15.6%
20-39
40-59
60-79
Age groups

"Pre-diabetes" - If the sum of these alterations ("Pre-diabetes") is considered by age group we find them in 6.7% in the 20 to 39 year age group, 26.5% in the 40 to 59 year age group and 46.2% in the 60 to 79 year age group. It can therefore be said that nearly half of the people in the oldest age group are in a pre-diabetes situation - 46.2%.

DIABETES AND "PRE-DIABETES"

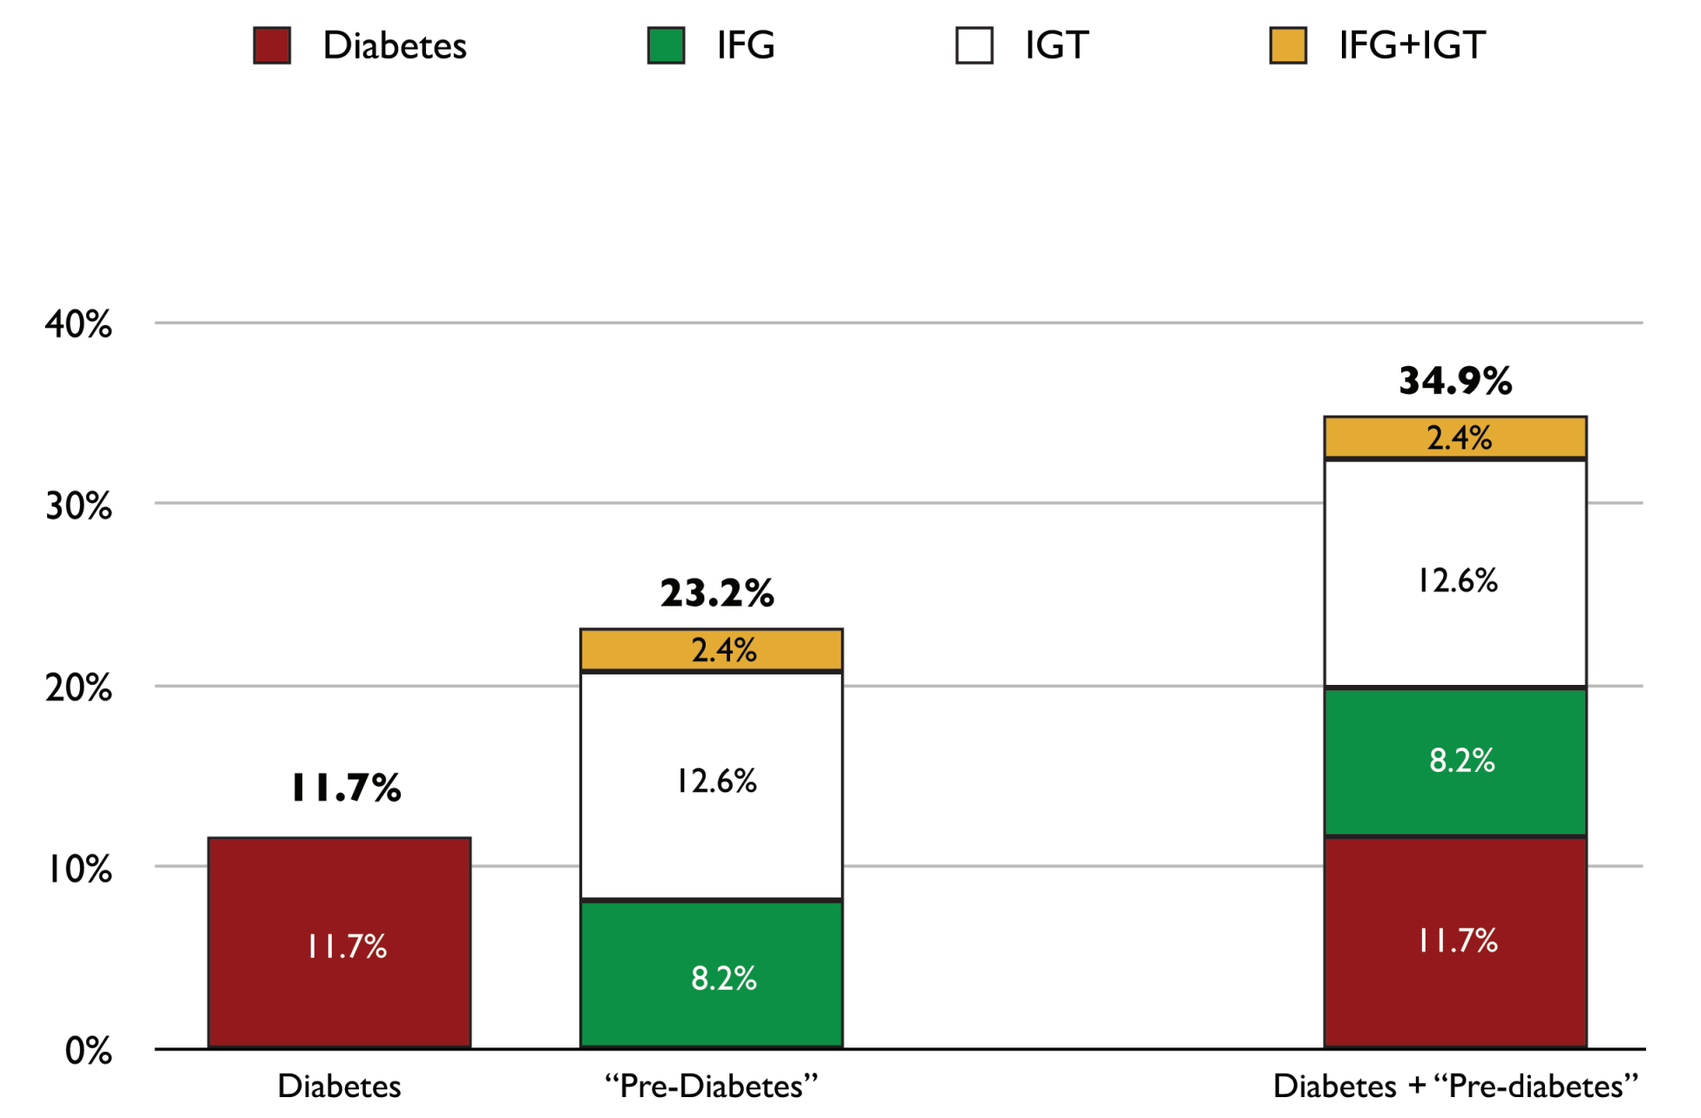
Diabetes
IFG
IGT
IFG+IGT
40%
30%
20%
10%
0%
11.7%
11.7%
23.2%
2.4%
12.6%
8.2%
34.9%
2.4%
12.6%
8.2%
11.7%
Diabetes
"Pre-Diabetes"
Diabetes + "Pre-diabetes"

Diabetes and "pre-diabetes" – 34.9% of the Portuguese population between the ages of 20 and 79 years present alterations in the carbohydrate metabolism.

A1C IN UNDIAGNOSED DIABETES (n=264)

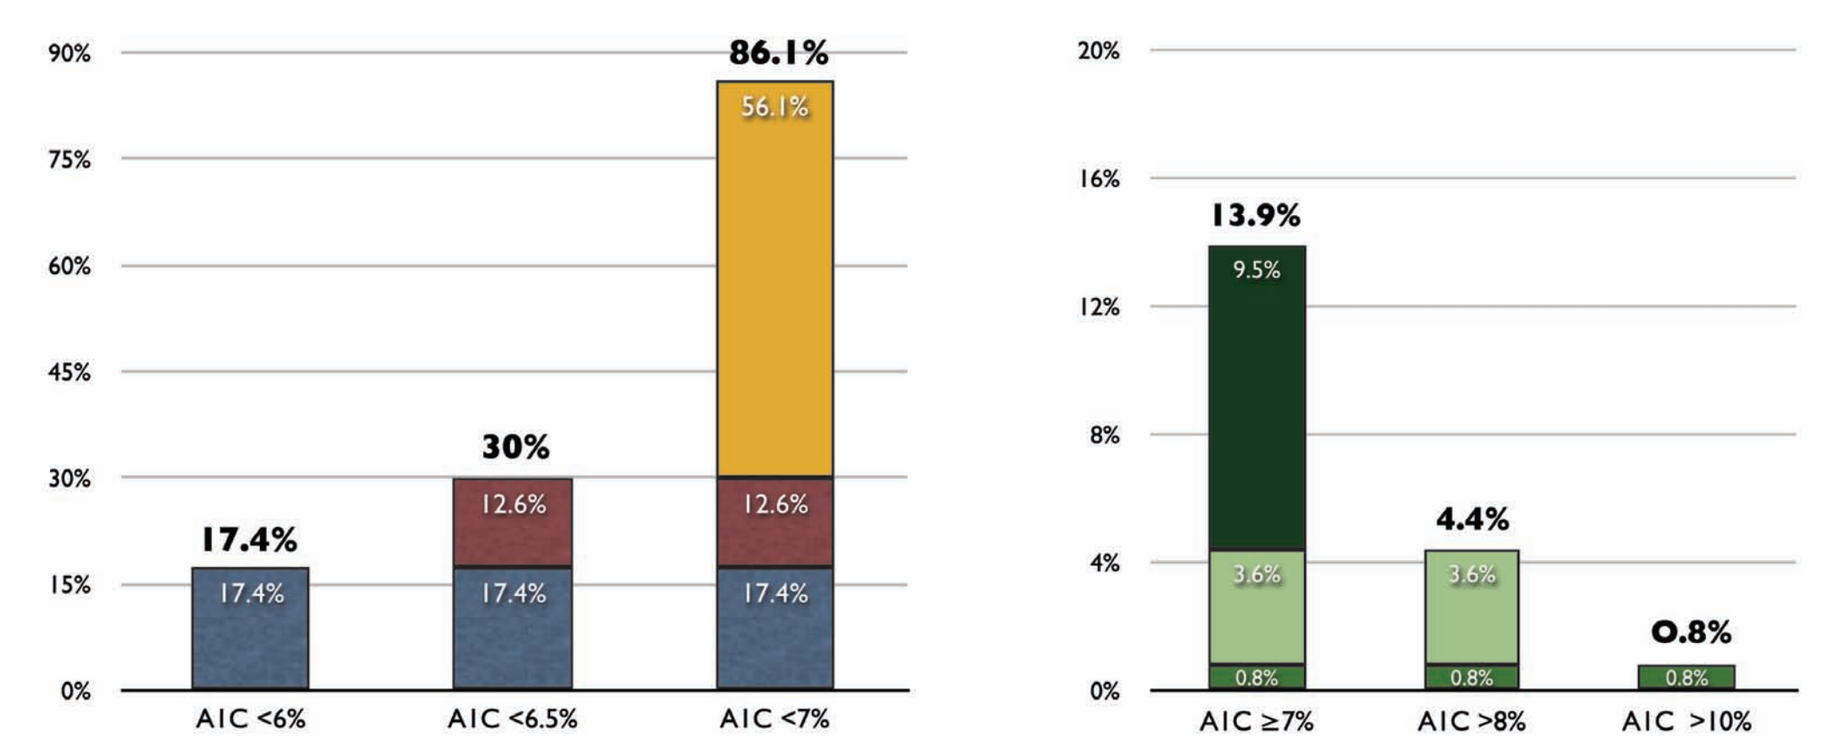
90%
75%
60%
45%
30%
15%
0%
17.4%
17.4%
30%
12.6%
17.4%
86.1%
56.1%
12.6%
17.4%
A1C <6%
A1C <6.5%
A1C <7%
20%
16%
12%
8%
4%
0%
13.9%
9.5%
3.6%
0.8%
4.4%
3.6%
0.8%
0.8%
0.8%
A1C ≥7%
A1C >8%
A1C >10%

A1C IN THE POPULATION WITHOUT PREVIOUS DIAGNOSIS OF DIABETES (n=4826)

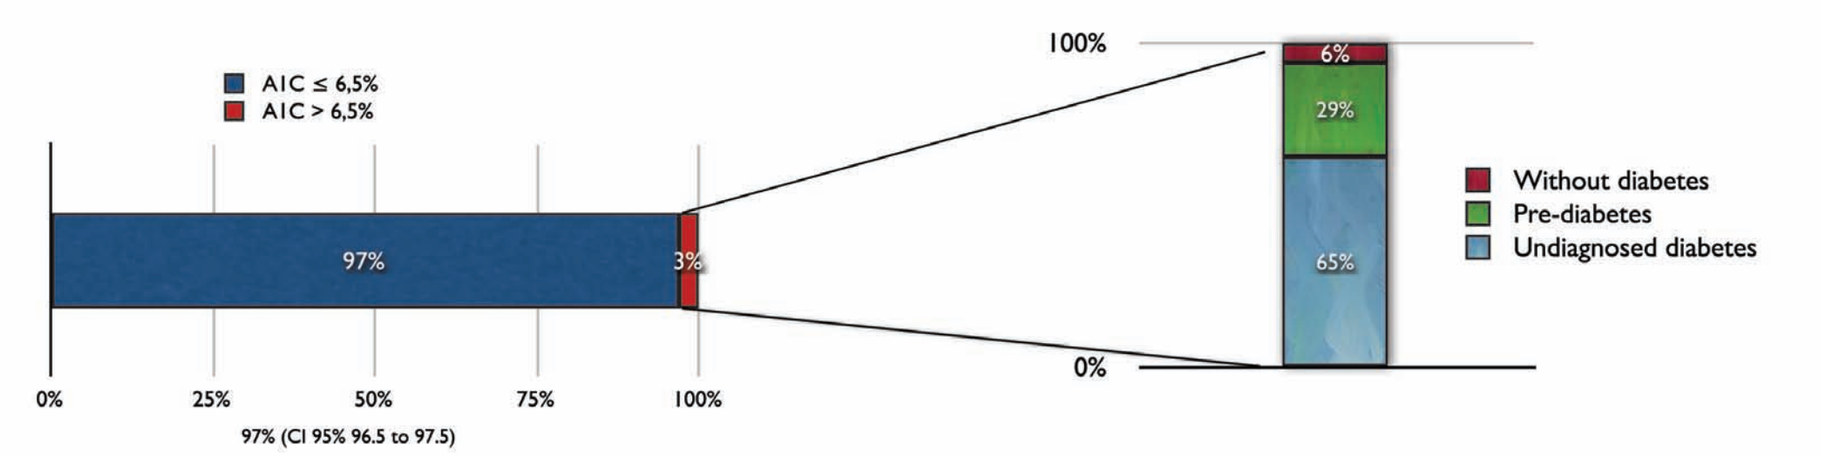
A1C ≤ 6,5%
A1C > 6,5%
97%
3%
0%
25%
50%
75%
100%
97% (CI 95% 96.5 to 97.5)
100%
0%
6%
29%
65%
Without diabetes
Pre-diabetes
Undiagnosed diabetes

BLOOD PRESSURE AND LDL-C IN THE DIABETIC POPULATION

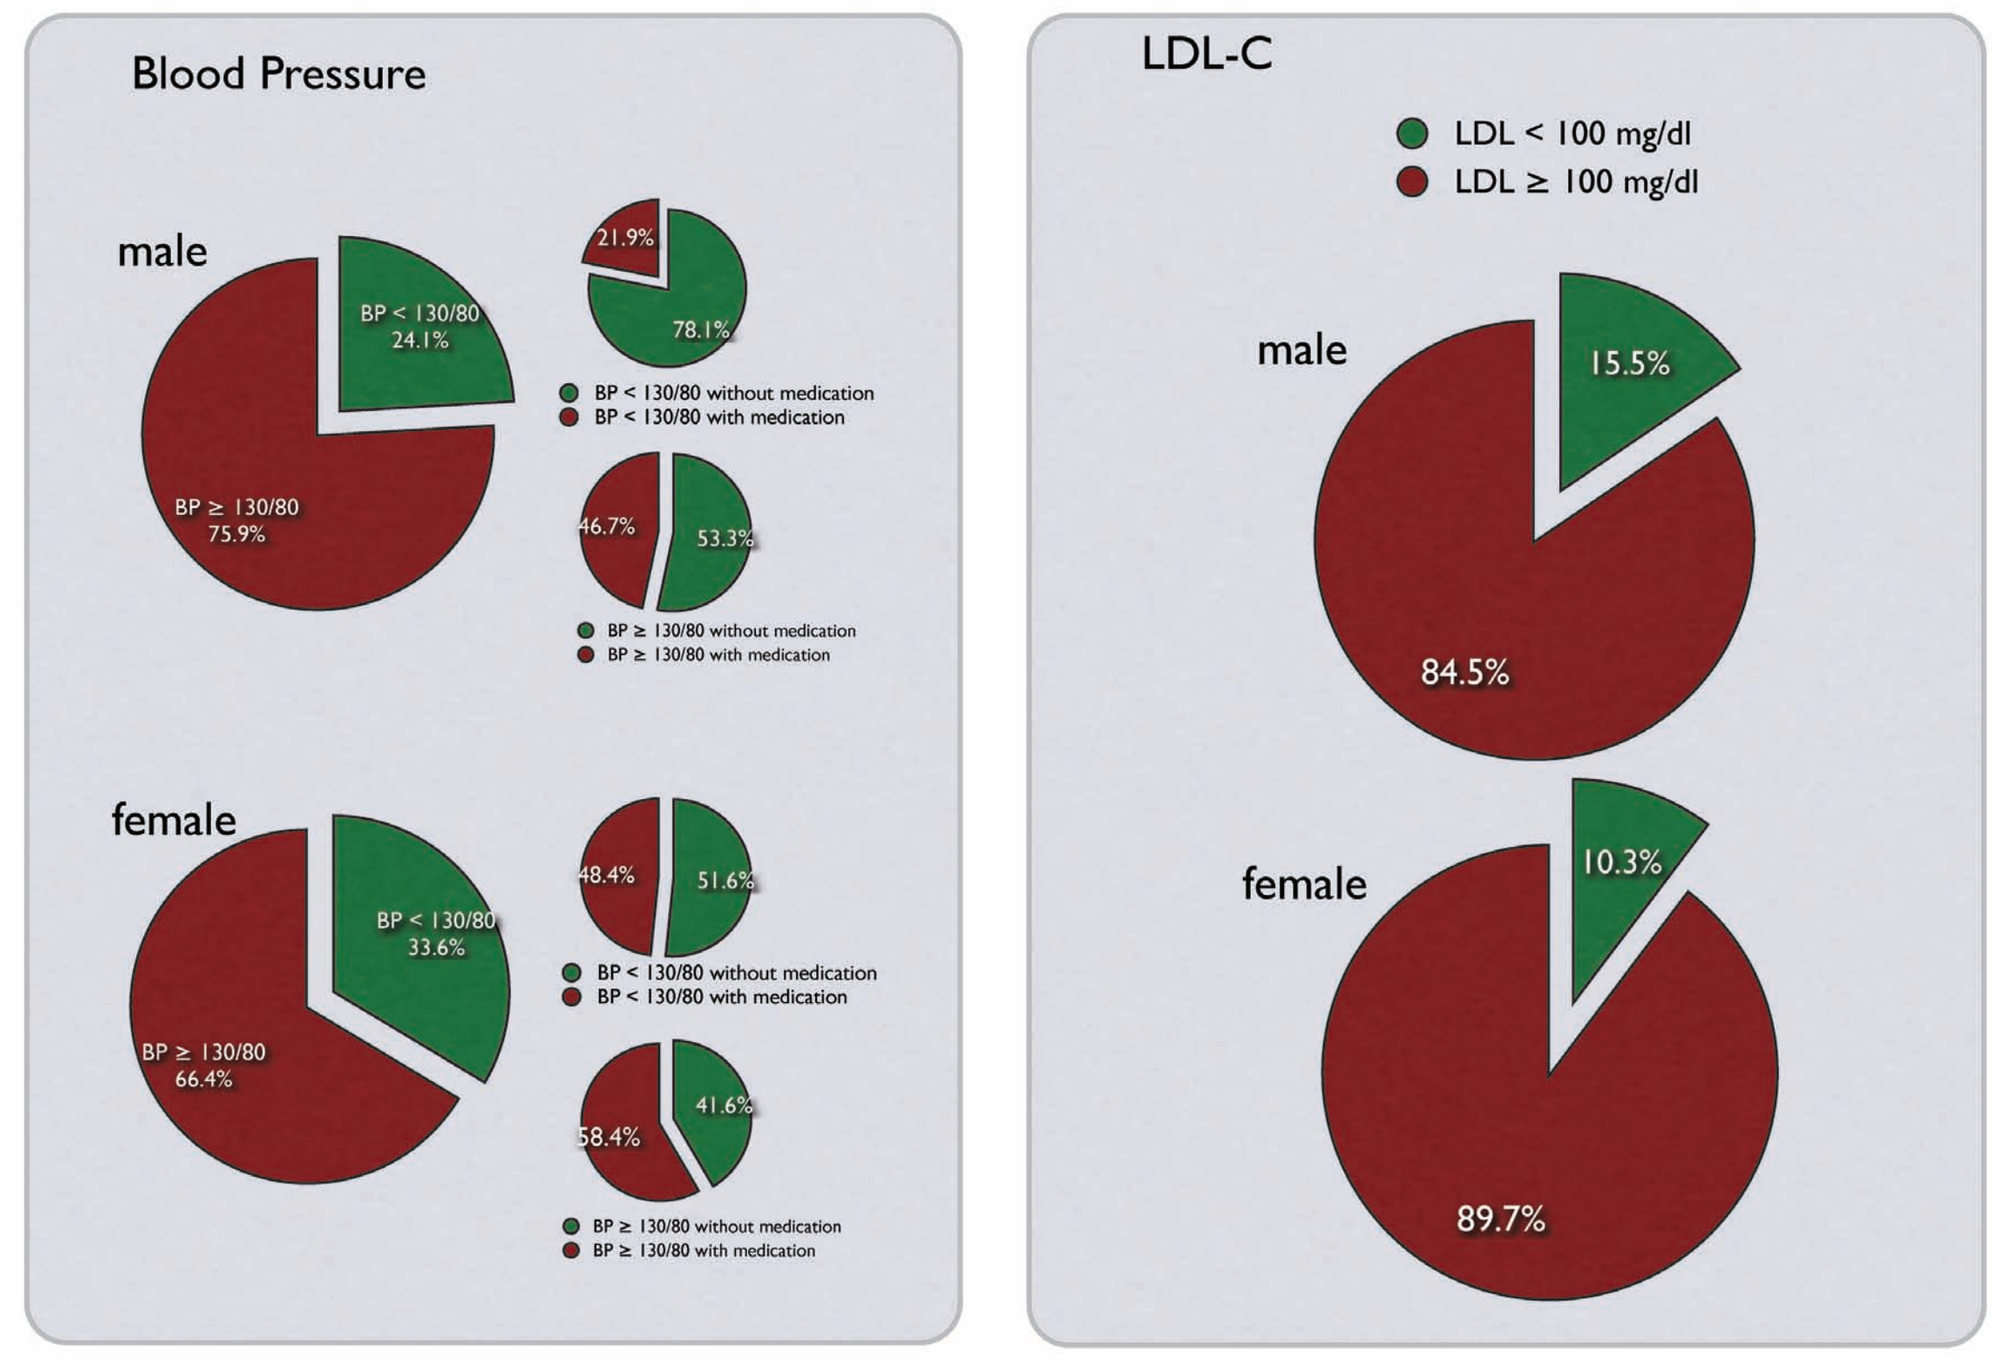
Blood Pressure
male
BP < 130/80
24.1%
BP ≥ 130/80
75.9%
21.9%
78.1%
BP < 130/80 without medication
BP < 130/80 with medication
46.7%
53.3%
BP ≥ 130/80 without medication
BP ≥ 130/80 with medication
female
BP < 130/80
33.6%
BP ≥ 130/80
66.4%
48.4%
51.6%
BP < 130/80 without medication
BP < 130/80 with medication
58.4%
41.6%
BP ≥ 130/80 without medication
BP ≥ 130/80 with medication
LDL-C
LDL < 100 mg/dl
LDL ≥ 100 mg/dl
male
15.5%
84.5%
female
10.3%
89.7%

CONCLUSIONS

- An 11.7% prevalence of diabetes was detected in the Portuguese population (20-79 years old).
- A significant difference was found between men (14.2%) and women (9.5%). Of these, 6.6% had previously been diagnosed and 5.1 % did not know they had diabetes.
- The total comparative prevalence of diabetes found was 9.7% (5.3% in diagnosed diabetes and 4.4% in undiagnosed diabetes).
- The study indicates that 43.6% of the diabetic population are unaware they have this chronic disease.
- 23.2% of persons aged between 20 and 79 years, were found to have "pre-diabetes" which is associated to the risk of diabetes and cardiovascular disease. 34.9% of the Portuguese population between the ages of 20 and 79 years suffer from diabetes or "pre-diabetes".
- Unknown diabetes was diagnosed by fasting glycaemia in 69,2% of the cases and in 30,8% by glucose tolerance test.
- Hypertension (TA ≥ 130/80 mmhg) was detected in 70.9% in both sexes.
- LDL Cholesterol ≥ 100 mg/dl was found in 85.7% of diabetics.

ACKNOWLEDGEMENTS

- This Study received a grant from the Portuguese Directorate General of Health.
- The study was only possible thanks to the hard work and dedication of the many doctors and nurses in each selected area. We are particularly grateful to the staff of the Portuguese Diabetes Association and to the regional heads of the National Diabetes Program who ensured that appropriate conditions would be provided for the project locally.
- No potential conflicts of interest relevant to this article were reported.

Ministério da Saúde
Direcção-Geral da Saúde

REFERENCES

1. American Diabetes Association: Diagnosis and classification of diabetes mellitus. Diabetes Care 31 (Suppl. 1): S55-S60, 2008
2. American Diabetes Association: Economic costs of diabetes in the U.S. in 2007. Diabetes Care 31:596-615, 2008
3. Amos AF, McCarty DJ, Zimmet P: The rising global burden of diabetes and its complications: estimates and projections to the year 2010. Diabet Med 1
4. (Suppl. 5): S1-S85, 19974.Catherine C. Cowie, PHD, Keith F. Rust, PHD, Earl S. Ford, MD, Mark S. Eberhardt, PHD, Danita D. Byrd-Holt, Chaoyang Li, MD, Desmond E. Williams, MD, Edward W. Gregg, PHD, Kathellen E. Bainbridge, PHD, Sharon H. Saydah, PHD, Linda S. Geiss, MS: Full Accounting of Diabetes and Pre-Diabetes in the U.S. Population in 1988-1994 and 2005-2006
5. Centers for Disease Control and Prevention National Center for Health Statistics: National Health and Nutrition Examination Survey 2005-2006 [Internet], 2008 . Available from: http://www.cdc.gov./nchs/about/major/nhames/nhames2005-2006/nhames05_06.htm. Accessed 28 May 2008
6. Greg EW, Cadwell BL, Chang YJ, Cowie CC, Williams DE, Geiss L, Engelgau MM, Vinicor F: Trends in the prevalence and ratio of diagnosed to undiagnosed diabetes according to obesity levels in the U.S. Diabetes Care 27:2806-2812, 2004
7. Ilhan Satman, MD, Temel Yilmaz, MD, Ahmet Sengül, MD, Serpil Salman, MD, Fatih Salman , MD, Sevil Uygur, Irfan Bastar, PHD, Yildiz Tütüncü, Mehmet Sargin, MD, Nevin Dinççag, MD, Kubilay Karsidag, MD, Sibel Kalaça, MD, Cihangir Özcan, MD, Hilary King, MD, DSc, The TURDEP Group: Population-Based Study of Diabetes and Risk Characteris tics in Turkey – Results of the Turkish Diabetes Epidemiology Study (TURDEP)
8. International Diabetes Federation: Diabetes Atlas 2006. Brussels, International Diabetes Federation, 2006
9. King H, Aubert RE, Herman WH: Global burden of diabetes, 1995-2025: prevalence, numerical estimates, and projections. Diabetes Care 21:1414-1431, 1998
10. King H, Rewers M: Global estimates for prevalence of diabetes mellitus and impaired glucose tolerance in adults: WHO Ad Hoc Diabetes Reporting Group. Diabetes Care 16:157-177, 1993
11. Knowler WC, Barrett-Connor E, Fowler SE, Hamman RF, Lachin JM, Walker EA, Nathan DM: Reduction in the incidence of type 2 diabetes with lifestyle intervention or metformin. N Engl J Med 346:393-403, 2002
12. Sarah Wild, MB Bchir, PHD, Gojka Roglic, MD, Anders Green, MD, PHD, DR. MED SCI, Richard Sicree, MBBS, MPH, Hilary King, MD, DSC: Global Prevalence of Diabetes – Estimates for the year 2000 and projections for 2030
13. Satman I, Yilmaz T, Sengul A, Salman S, Salman F, Uygur S, Bastar I, Tutuncu Y, Sargin M, Dinccag N, Karsidag K, Kalaca S, Ozcan C, King H: Population-based study of diabetes and risk characteristics in Turkey: results of the Turkish Diabetes Epidemiology Study (TURDEP). Diabetes Care 25:1551-1556, 2002
14. Wild S, Roglic G, Green A, Sicree R, King H, Global prevalence of diabetes: estimates for the year 2000 and projections for 2030. Diabetes Care 2004;24:1047-53
15. Inquérito Nacional de Saúde 2005/2006", National Health Institute/National Health Inquire, Lisboa, 2006 - 37-42
16. International Expert Committee Report on the Role of the A1C Assay in the Diagnosis of Diabetes, The International Expert Committee. David M. Nathan, MD (Chair); Beverly Balkau, PhD; Enzo Bonora, MD, PhD; Knut Borch-Johnsen, MD, DMSc; John B. Buse, MD, PhD; Stephen Colagiuri, MD; Mayer B. Davidson, MD; Ralph DeFronzo, MD; Saul Genuth, MD; Rury R. Holman, FRCP; Linong Ji, MD; Sue Kirkman, MD; William C. Knowler, MD, Dr PH; Desmond Schatz, MD; Jonathan Shaw, MD; Eugene Sobngwi,MD; Michael Steffes, MD, PhD; Olga Vaccaro, MD; Nick Wareham, MD; Bernard Zinman, MD; and Richard Kahn, PhD. Diabetes Care, 32, 7: 1-8, 2009.